1352

A Phase 3 open-label trial-in-progress evaluating garadacimab for prophylactic treatment of pediatric patients (aged 2–11 years) with hereditary angioedema

Andrea Zanichelli,[1,2] Avner Reshef,[3] Raffi Tachdjian,[4] William R. Lumry,[5] Inmaculada Martinez Saguer,[6] Markus Magerl,[7–9] Mauro Cancian,[10] Michael E. Manning,[11] William H. Yang,[12] Jonathan A. Bernstein,[13] Connie Hsu,[14] Lolis Wieman,[15] Emel Aygören-Pürsün[16]

[1]Operative Unit of Medicine, Angioedema Center, IRCCS Policlinico San Donato, San Donato Milanese, Milan, Italy; [2]Department of Biomedical Sciences for Health, University of Milan, Milan, Italy; [3]Allergy, Immunology and Angioedema Center, Barzilai University Hospital, Ashkelon, Israel; [4]Division of Allergy and Clinical Immunology, David Geffen School of Medicine at University of California Los Angeles, Los Angeles, CA, USA; [5]AARA Research Center, Dallas, TX, USA; [6]HZRM Haemophilia Center Rhein Main, Mörfelden-Walldorf, Germany; [7]Institute of Allergology, Charité–Universitätsmedizin Berlin, Berlin, Germany; [8]Corporate Member of Freie Universität Berlin and Humboldt-Universität zu Berlin, Berlin, Germany; [9]Fraunhofer Institute for Translational Medicine and Pharmacology ITMP, Immunology and Allergology, Berlin, Germany; [10]UOSD Allergologia, University Hospital of Padua, Padua, Italy; [11]Allergy, Asthma & Immunology Associates, Ltd; Internal Medicine, UA College of Medicine – Phoenix, Phoenix, AZ, USA; [12]Ottawa Allergy Research Corporation; Department of Medicine, University of Ottawa, Ottawa, ON, Canada; [13]University of Cincinnati, Department of Internal Medicine Division of Rheumatology, Allergy and Immunology, and the Bernstein Clinical Research Center Cincinnati, Cincinnati, OH, USA; [14]Research Solutions of Arizona, PC, Litchfield Park, AZ, USA; [15]CSL Behring, King of Prussia, PA, USA; [16]University Hospital Frankfurt, Department for Children and Adolescents, Goethe University Frankfurt, Frankfurt, Germany.

CONCLUSIONS

- **This multicenter, open-label, single arm, Phase 3 study (NCT05819775) will provide garadacimab data on safety, efficacy, and pharmacokinetics/pharmacodynamics in pediatric patients with hereditary angioedema (HAE)**
- **This study will aim to address the existing unmet need for novel prophylactic treatment in the pediatric population**
- **Recruitment was initiated in May 2023 with an estimated study completion date of November 2026**

BACKGROUND

HAE[1,2]

- HAE causes recurrent, unpredictable and potentially life-threatening episodes of swelling (attacks)
- Bradykinin is the key mediator of HAE attacks
- HAE symptom onset usually occurs in the first or second decade of life

Garadacimab[3,4]

- First-in-class, fully-human monoclonal antibody targeting activated factor XII (FXIIa)
- FXIIa is a key initiator of the contact system, upstream of bradykinin production
- High affinity/potency/specificity; decreases bradykinin production *in vitro*
- Under clinical evaluation in **pivotal Phase 3 (VANGUARD)** and **Phase 3 open-label extension (OLE)** studies

GARADACIMAB ADOLESCENT SUBGROUP DATA GENERALLY CONSISTENT WITH OVERALL POPULATION IN PIVOTAL PHASE 3 AND OLE STUDIES

Durable efficacy and favorable long-term safety profile in adults and adolescents with garadacimab 200 mg subcutaneous once-monthly[4–7]

Overall population (aged ≥12 years)

Pivotal Phase 3 study[4]
(6-month treatment period)

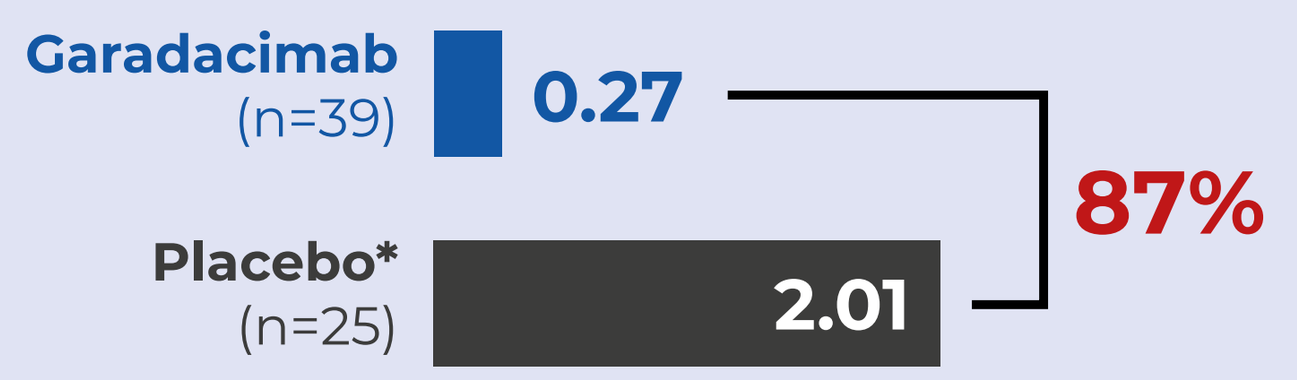
Garadacimab
(n=39)
0.27
Placebo*
(n=25)
2.01
87%

87% reduction in mean monthly number of HAE attacks vs placebo

OLE study[5]
(median exposure 13.8 months)

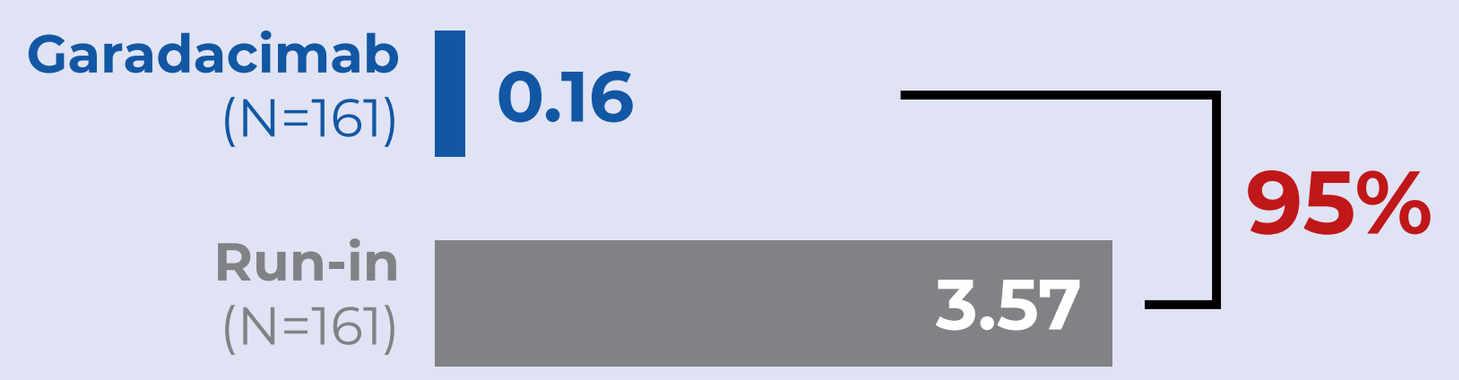
Garadacimab
(N=161)
0.16
Run-in
(N=161)
3.57
95%

95% reduction in mean monthly number of HAE attacks vs run-in

Pivotal Phase 3 (garadacimab n=39) and OLE (N=159) safety profile:[4,5]

- No deaths
- No discontinuations due to TEAEs in pivotal Phase 3 study; one discontinuation in OLE†
- One SAE‡ in pivotal Phase 3 study and three SAEs§ in OLE study; none related to garadacimab
- No AESIs per protocol¶

Adolescent subgroup (aged 12 to ≤17 years)

Pivotal Phase 3 study in adolescent subgroup[6]
(6-month treatment period)

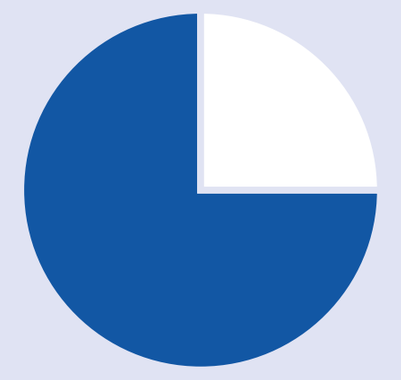

75% (3/4) of adolescents achieved ≥90% reduction in mean monthly number of HAE attacks vs run-in

OLE study in adolescent subgroup[7]
(median exposure 13.8 months)

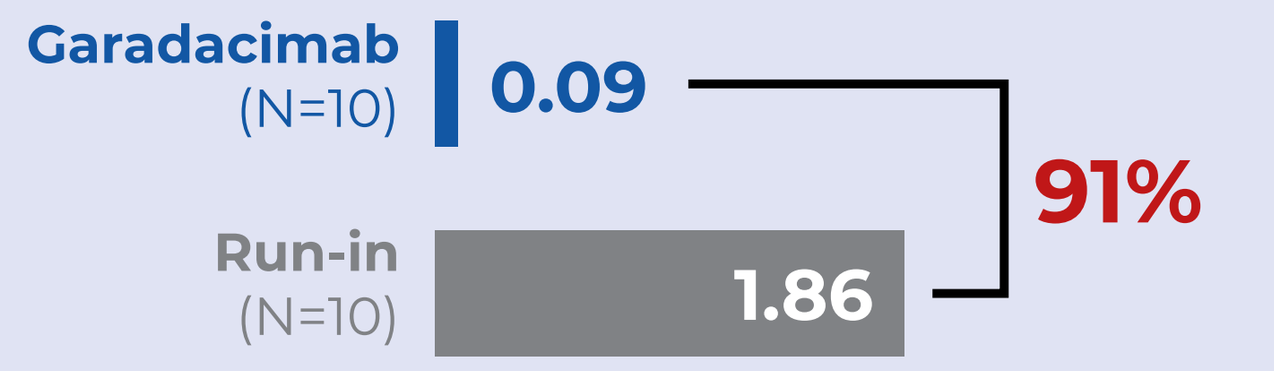
Garadacimab
(N=10)
0.09
Run-in
(N=10)
1.86
91%

91% reduction in mean monthly number of HAE attacks vs run-in

Pivotal Phase 3 (garadacimab, n=4) and OLE (N=10) safety profile in adolescents:[6,7]

- No deaths due to TEAEs
- No discontinuations due to TEAEs
- No SAEs
- No AESIs per protocol¶
- No garadacimab-related TEAEs

Pivotal Phase 3 (VANGUARD) study: During the 6-month treatment period, the mean monthly number of HAE attacks (primary endpoint) was significantly lower in the garadacimab group (0.27, 95% CI 0.05–0.49) than in the placebo group* (2.01, 1.44–2.57; P<0.0001), the median monthly number of HAE attacks was 0 (IQR 0.00–0.31) for garadacimab and 1.35 (1.00–3.20) for placebo.
OLE study (median exposure 13.8 months): The mean monthly number of HAE attacks (secondary endpoint) was lower with garadacimab (0.16) versus run-in (3.57). Similar results were found in the adolescent subgroup where the mean monthly number of HAE attacks was lower with garadacimab (0.09) versus run-in (1.86).
*One patient in the placebo group was excluded from the efficacy analysis as they received treatment for <30 days; †One garadacimab-related ISR leading to study discontinuation; ‡One severe SAE (laryngeal attack) was assessed as not related to trial treatment: The patient made a full recovery and was kept under hospital observation overnight; §COVID-19, n=2; abdominal HAE attack, n=1; ¶As defined in the protocol, AESIs included severe hypersensitivity including anaphylaxis, thromboembolic, or abnormal bleeding events. None of the events met AESI criteria as per protocol.
AESI, adverse event of special interest; CI, confidence interval; HAE, hereditary angioedema; IQR, interquartile range; ISR, injection-site reaction; OLE, open-label extension; SAE, serious adverse event; TEAE, treatment-emergent adverse event.

MULTICENTER, OPEN-LABEL, SINGLE-ARM, PHASE 3 STUDY (NCT05819775) WITH GARADACIMAB IN PEDIATRIC PATIENTS WITH HAE[8]

GOAL: To address existing unmet need for effective, well-tolerated, and convenient treatment options for pediatric patients with HAE

Screening (up to 1 month)

Inclusion criteria
- Diagnosed with HAE-C1-INH
- Aged between 2 and 11 years*
- ≥2 attacks in last 6 months

Exclusion criteria
- Concomitant diagnosis with other forms of angioedema
- Pre-planned major surgeries or procedures
- HAE treatments within minimum 2 weeks before treatment period†
- Participation in another interventional clinical study

Treatment (12 months)

Garadacimab Dose A SC
Patients aged 2–5 years

Garadacimab Dose B SC
Patients aged 6–11 years

Follow-up (3 months after last dose)

*With body weight ≥10th percentile based on age, at the time of providing written informed consent; †Use of C1-INH products, androgens, antifibrinolytics, approved or future approved medications, or other small molecule medications for routine prophylaxis against HAE attacks within 2 weeks of study treatment.
C1-INH, C1-esterase inhibitor; HAE, hereditary angioedema; HAE-C1-INH, hereditary angioedema with C1-inhibitor deficiency or dysfunction; SC, subcutaneous.

Primary safety and PK endpoints

TEAEs
- Number and percentage of patients with TEAEs
- Number of events
- TEAE rates per injection
- TEAE rates per patient-year

PK parameters at steady state
- C_{max}
- C_{trough}
- T_{max}

Secondary efficacy endpoints

- Time-normalized number of attacks
- Time-normalized number of attacks requiring on-demand treatment
- Time-normalized number of moderate and/or severe attacks
- Percentage reduction in the time-normalized number of attacks

Recruitment is planned across global sites

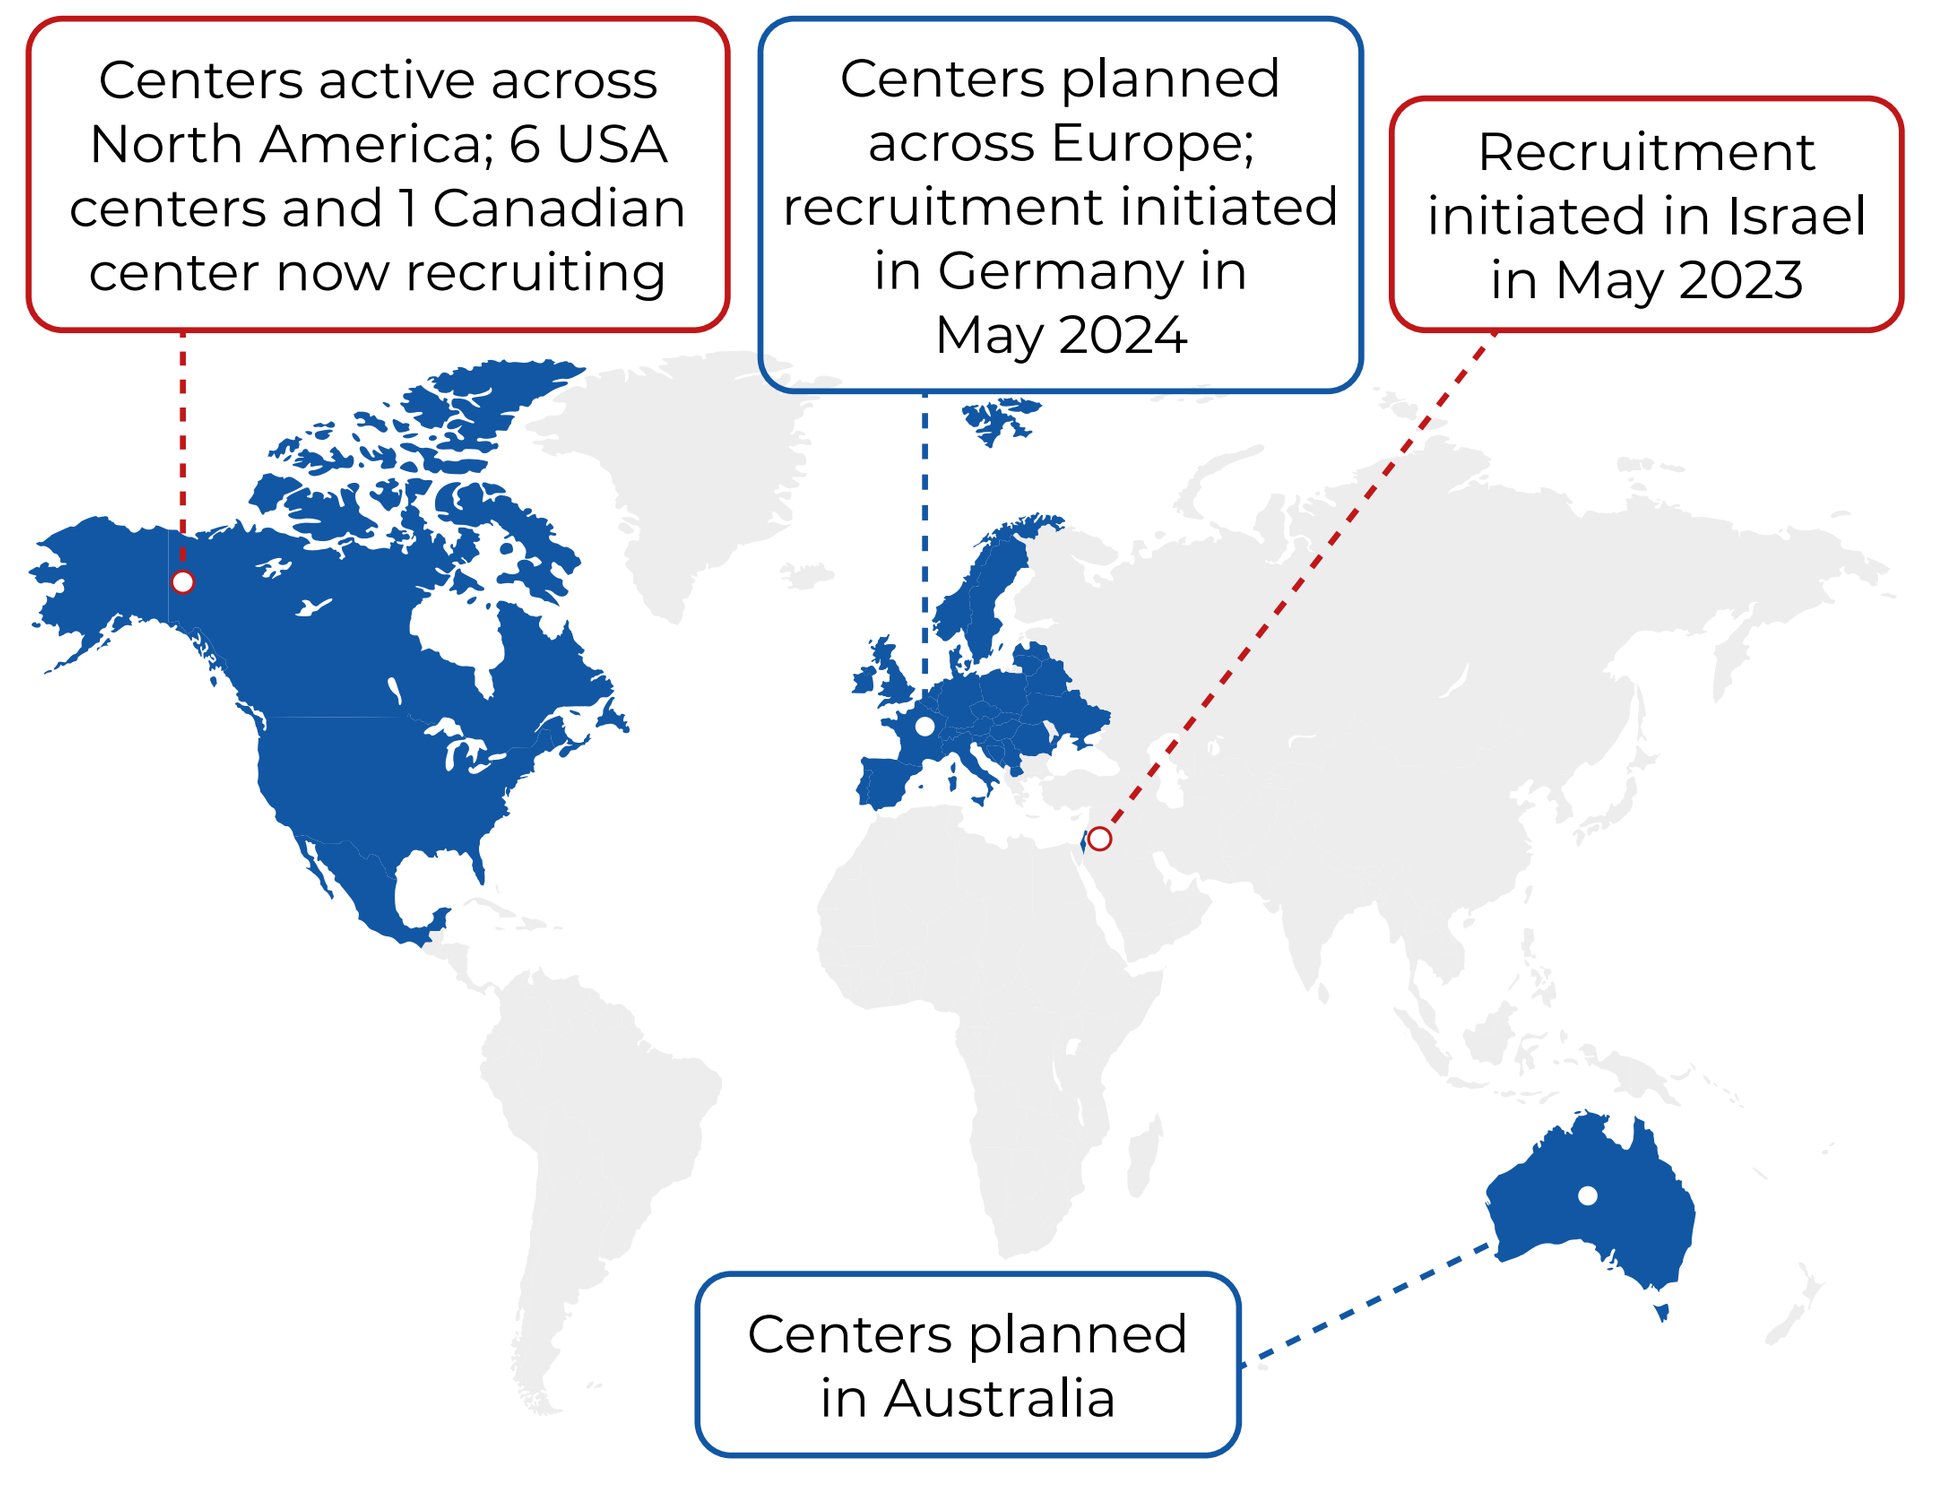
Centers active across North America; 6 USA centers and 1 Canadian center now recruiting
Centers planned across Europe; recruitment initiated in Germany in May 2024
Recruitment initiated in Israel in May 2023
Centers planned in Australia

Target enrollment is 20 patients

- **Ten patients screened and eight patients receiving treatment**
- **First enrolled patient has been in the study for 1 year**

C_{max}, maximum concentration; C_{trough}, trough concentration at steady state; PK, pharmacokinetics; TEAE, treatment-emergent adverse event; T_{max}, time to reach C_{max} in plasma.

Conflicts of interest:
A. Zanichelli has received honoraria as a speaker/advisor from BioCryst, BioMarin, CSL Behring, KalVista, Pharming, Pharvaris, and Takeda/Shire.

References
1. Busse P et al. *J Allergy Clin Immunol Pract* 2021;9:132–150.e3; 2. MacGinnitie JA. *Pediatr Allergy Immunol* 2014;25:420–427; 3. Cao H et al. *J Allergy Clin Immunol* 2018;142:1355–1358; 4. Craig TJ et al. *Lancet* 2023;401:1079–1090; 5. Anderson J et al. Abstract A016. Oral presented at the American College of Allergy, Asthma & Immunology 2023 Annual Scientific Meeting, November 9–13, 2023, Anaheim, California; 6. Magerl M et al. Poster 48. Poster presented at the at the 13th C1-inhibitor Deficiency & Angioedema Workshop, May 6 2023, Budapest, Hungary; 7. Jacobs JS et al. Poster 016. Poster presented at the American Academy of Allergy, Asthma, and Immunology Annual Meeting, February 23–26 2024, Washington DC, USA; 8. ClinicalTrials.gov. NCT05819775. Available at: https://classic.clinicaltrials.gov/ct2/show/NCT05819775 (accessed May 2024).

Disclosures:
Medical writing support was provided by Helix, OPEN Health Communications, and funded by CSL Behring, in accordance with Good Publication Practice (GPP) guidelines (www.ismpp.org/gpp-2022).

Scan to access the poster

European Academy of Allergy and Clinical Immunology Congress | Valencia, Spain | May 31 – June 3, 2024